San Antonio Breast Cancer Symposium® - December 8-11, 2020

Targeting Resistance to Current CDK4/6 Therapies by RGT-419B, an Inhibitor with an Optimized Kinase Activity Spectrum

Julie Xie[1], Jing Han[2], Zhilong Hu[2], Hu He[2], Xianqiang Sun[2], Fei Zhang[2], Xinjuan Wang[2], Liufeng Mei[2], Yangyang Liu[2], Xing Tong[2], Yuanshu Zhou[2], Guoyun Zhu[2], Lili Yao[2], Jing Lin[2], Xi Chen[2], Xiaotian Zhu[1], Wenge Zhong[2] and RGT-419B Team [1]Regor Pharmaceuticals Inc. Boston, MA, USA. [2]Qilu Regor Therapeutics Inc. Shanghai, PRC.

ABSTRACT

Cyclin-dependent kinases (CDKs) 4/6 inhibitors are a powerful class of therapeutic drugs for the treatment of advanced metastatic breast cancer. However, the currently approved CDK4/6 inhibitors palbociclib, ribociclib and abemaciclib have dose-limiting toxicities that require treatment holidays or reductions to sub-optimal doses, thus limiting sustained full target inhibition.[1] The residual CDK4/6 activity, together with persistent signaling through CDK2/cyclin E are among key resistant mechanisms that can compromise full clinical benefit.[2-3] RGT-419B is a new generation CDK inhibitor with an optimized kinase activity spectrum that has been discovered by deploying Computer Accelerated Rational Design (CARD) technology platform. RGT-419B has potent sub-nM CDK4 activity with desired degrees of selectivity against kinases such as CDK6, CDK9 and GSK3β, aiming to enable full target engagement with an improved safety profile. Furthermore, single digit nM CDK2 kinase activity has been incorporated into the design of RGT-419B to combat Cyclin E/CDK2-driven resistance. In vitro, RGT-419B showed more robust activity against palbociclib-resistant ER+ breast cancer cells than abemaciclib. In ER+ T47D breast cancer cells with overexpression of Cyclin E1, RGT-419B exhibited better anti-proliferation activity than either abemaciclib or palbociclib. RGT-419B also demonstrated more durable in vivo tumor growth inhibition when compared with abemaciclib in an ER+ breast cancer xenograft model. The optimized kinase activity spectrum of RGT-419B provides an opportunity to treat ER+ breast cancer patients refractory to the existing CDK4/6 inhibitors as either a single agent or in combination with other therapies.

RESULTS

Abbreviation: Abema: abemaciclib; **Palbo**: palbociclib; **SERD**: selective estrogen receptor degrader

		RGT-419B	Abemaciclib	Palbociclib
Biochemical activity IC_{50} (nM)	CDK4/Cyclin D1	0.5	0.8	2
	CDK2/Cyclin E1	6	270	>3000
Kinase selectivity against CDK4 (Fold)	CDK6/Cyclin D3	16	8	1
	CDK9/Cyclin T1	50	24	234
	GSK3β	> 1000	40	> 500
	CDK7 /Cyclin H	304	> 500	/
Cellular activity IC_{50} (nM)	T47D phospho-Rb (S807/811) inhibition	8	8	19
	T47D anti-proliferation	14	10	28
Cellular selectivity CDK6/CDK4 (Fold)	MV4-11/JeKo-1 anti-proliferation (fold)	6	4	1

Table 1. Activity and selectivity of RGT-419B in biochemical and cellular assays. RGT-419B has sub-nM CDK4 and single digit nM CDK2 IC_{50}. It potently suppressed the phosphorylation of the retinoblastoma protein and proliferation of T47D cell. RGT-419B showed improved selectivity against CDK6 in biochemical and cellular assays when compared with abemaciclib and palbociclib.

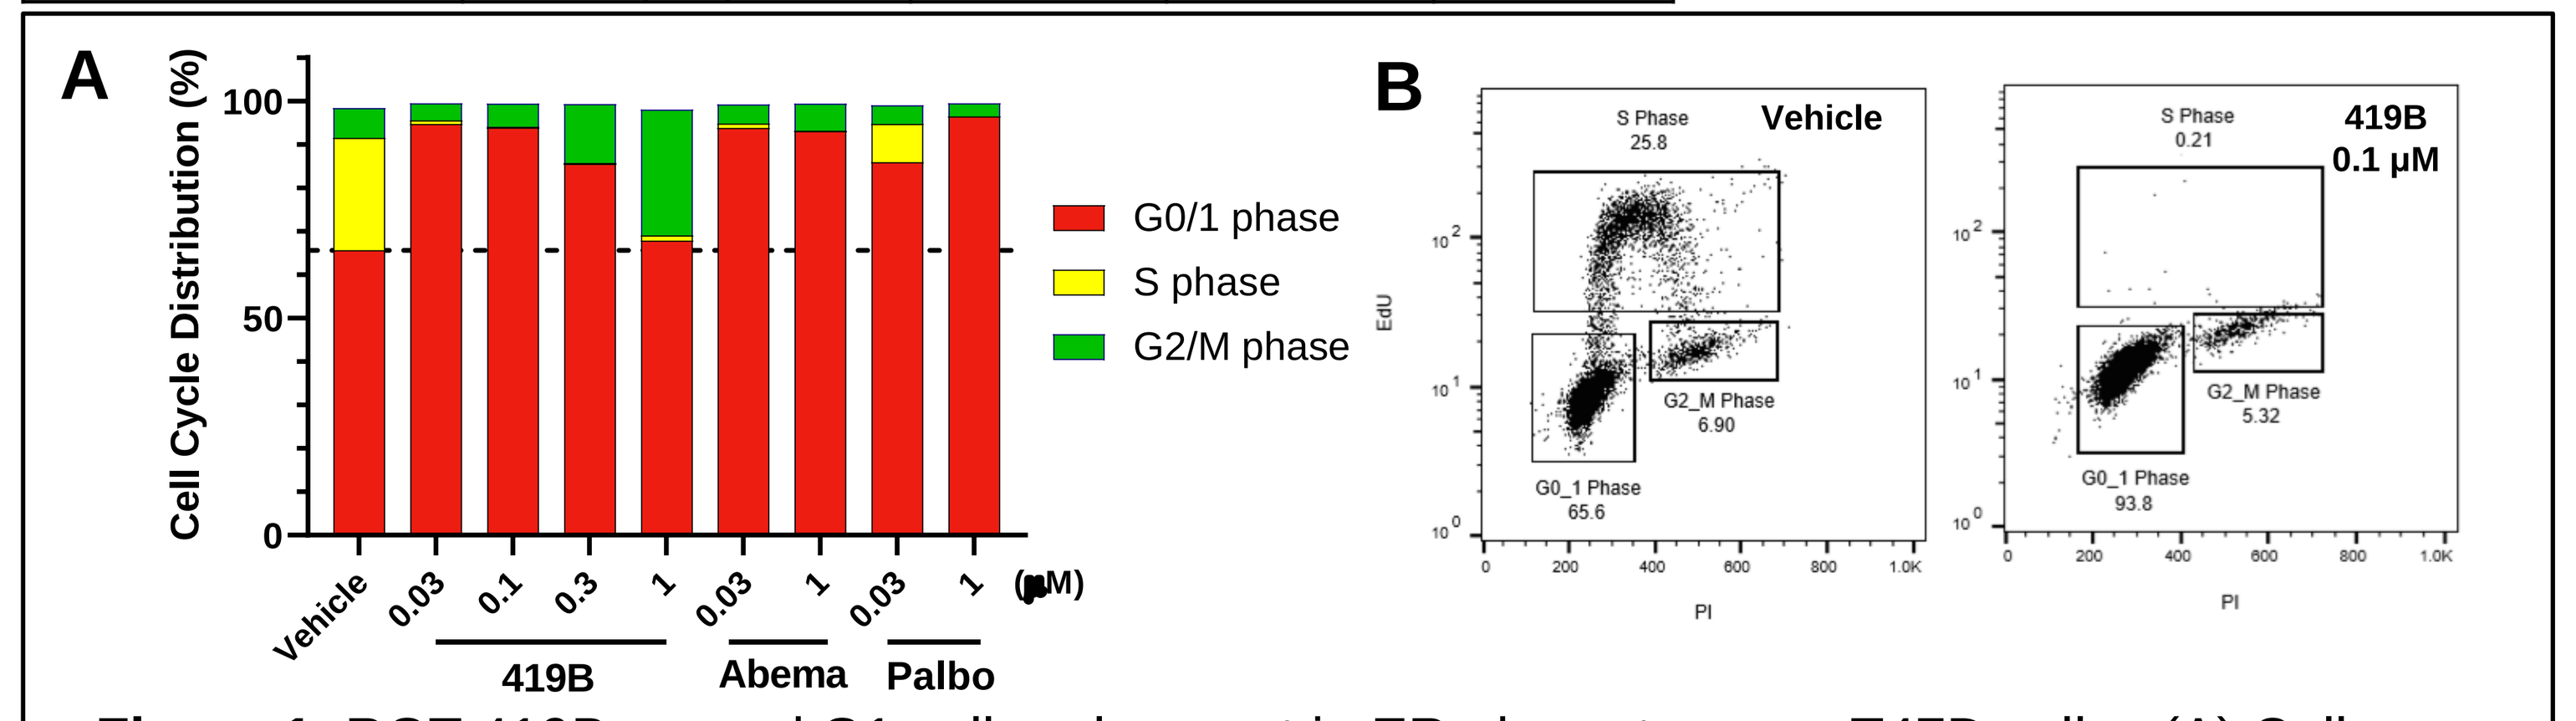
A
Cell Cycle Distribution (%)
G0/1 phase
S phase
G2/M phase
Vehicle
0.03
0.1
0.3
1
(μM)
419B
Abema
Palbo
B
Vehicle
419B 0.1 μM
S Phase
G2_M Phase
G0_1 Phase
EdU
PI

Figure 1. RGT-419B caused G1 cell cycle arrest in ER+ breast cancer T47D cells. (A) Cell cycle distribution of T47D cells was determined by EdU labeling after 24 hrs treatment with the indicated inhibitors. (B) Representative flow gating of Edu labeled and propidium iodide stained T47D cells treated with vehicle or RGT-419B at 0.1 μM.

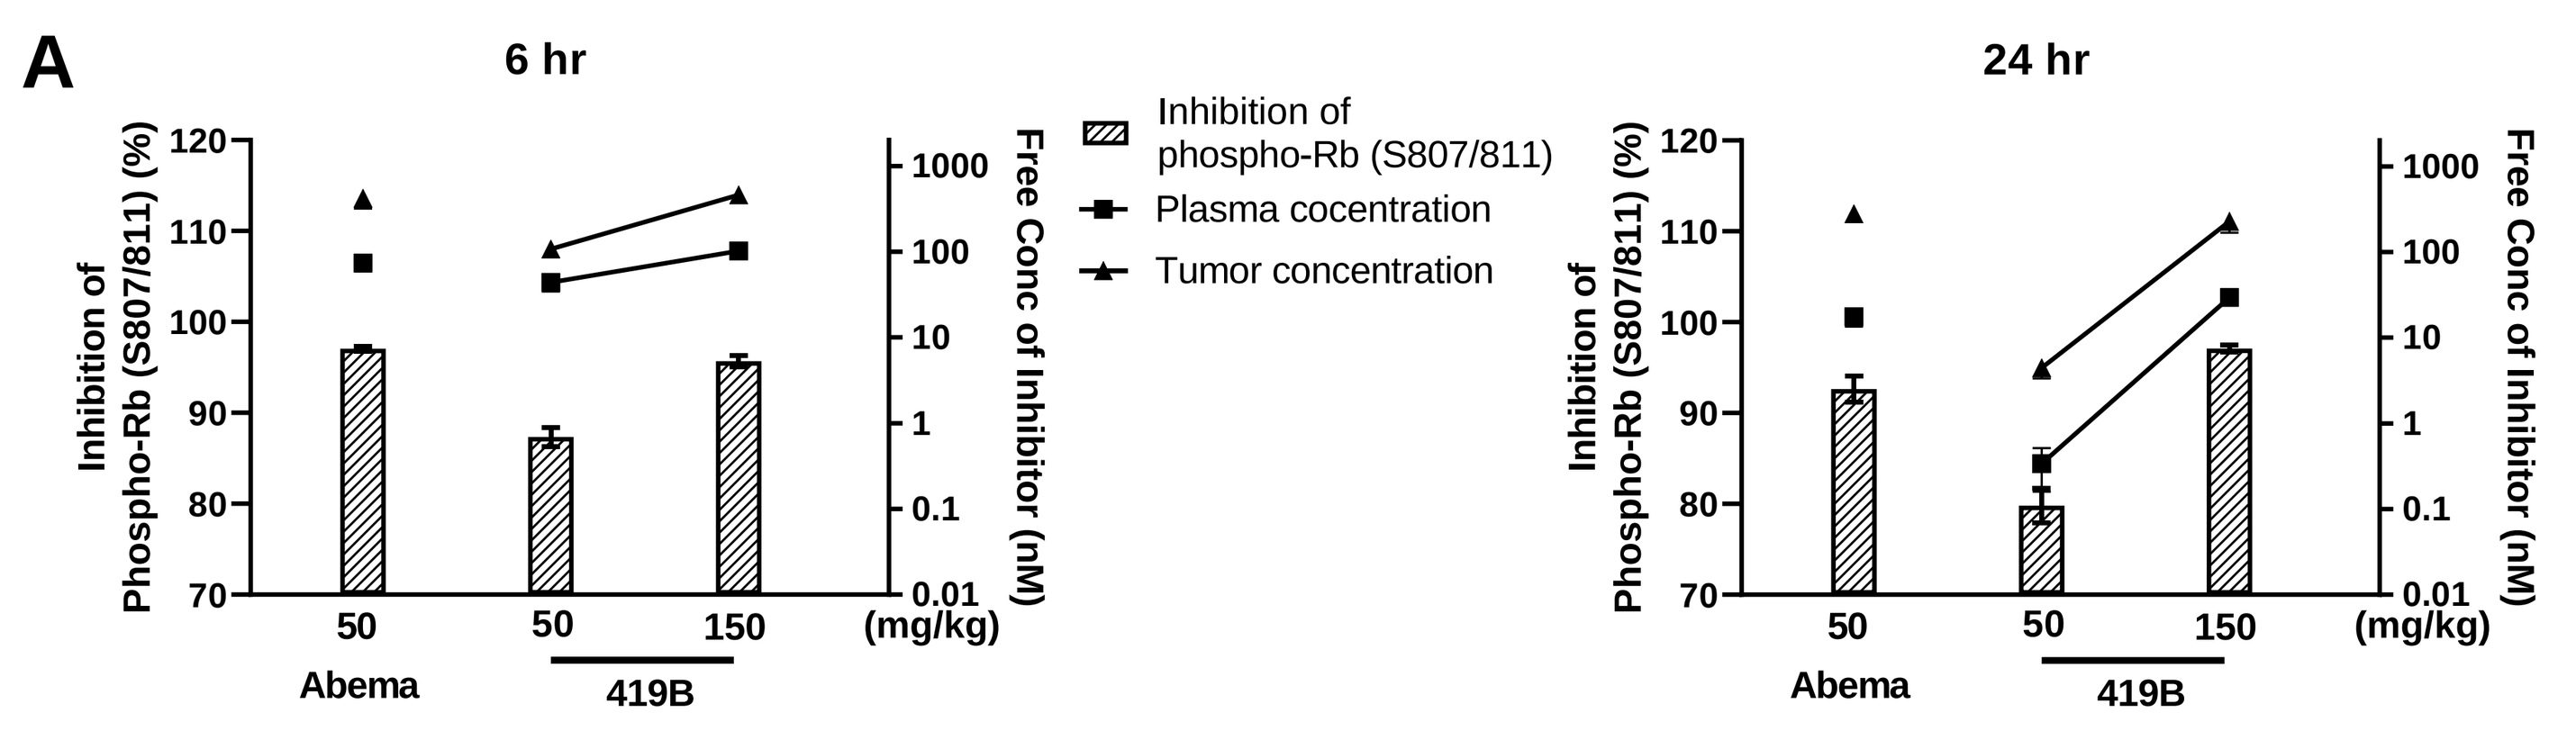
A
6 hr
24 hr
Inhibition of Phospho-Rb (S807/811) (%)
Free Conc of Inhibitor (nM)
Inhibition of phospho-Rb (S807/811)
Plasma cocentration
Tumor concentration
Abema
419B
(mg/kg)

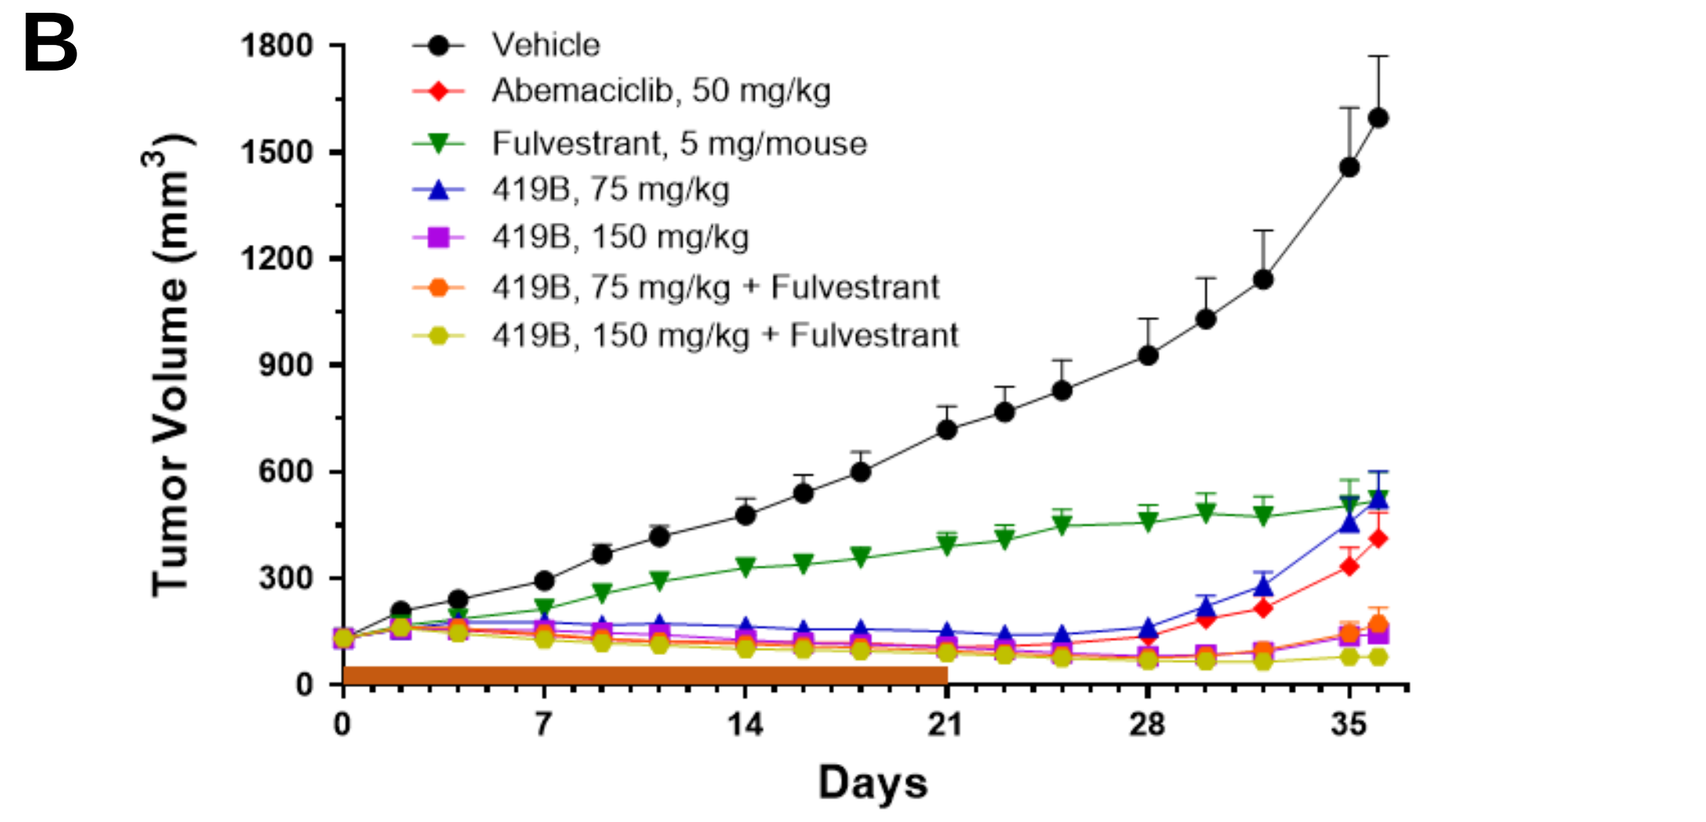
B
Tumor Volume (mm³)
Days
Vehicle
Abemaciclib, 50 mg/kg
Fulvestrant, 5 mg/mouse
419B, 75 mg/kg
419B, 150 mg/kg
419B, 75 mg/kg + Fulvestrant
419B, 150 mg/kg + Fulvestrant

	Body weight change on day 21 (%)
Vehicle	4.0
Abemaciclib, 50 mg/kg	-6.0
Fulvestrant, 5 mg/mouse	4.0
RGT-419B, 75 mg/kg	2.0
RGT-419B, 150 mg/kg	-9.0
RGT-419B, 75 mg/kg + Fulvestrant	2.0
RGT-419B, 150 mg/kg + Fulvestrant	-6.0

Figure 2. RGT-419B at 150 mpk demonstrated sustained tumor growth inhibition in ER+ breast cancer MCF7 xenograft model as single agent. (A) PK/PD of abemaciclib and RGT-419B in MCF7 xenograft model. The tumor bearing mice were treated with the indicated inhibitors once daily by oral gavage for 3 days. Plasma and tumors were collected at 6 and 24 hrs after the last dose at day 3 for the indicated measurements. Inhibition of phospho-Rb (S807/811) was calculated after normalization to β-actin. (B) Tumor growth inhibition efficacy in the ER+ MCF7 breast cancer xenograft. Orange bar in the X-axis represents the inhibitor treatment duration and the inhibitors were withdrawn after 21 days of treatment. RGT-419B and abemaciclib were dosed once daily through oral gavage and fulvestrant was dosed weekly at 5 mg via intraperitoneal injection.

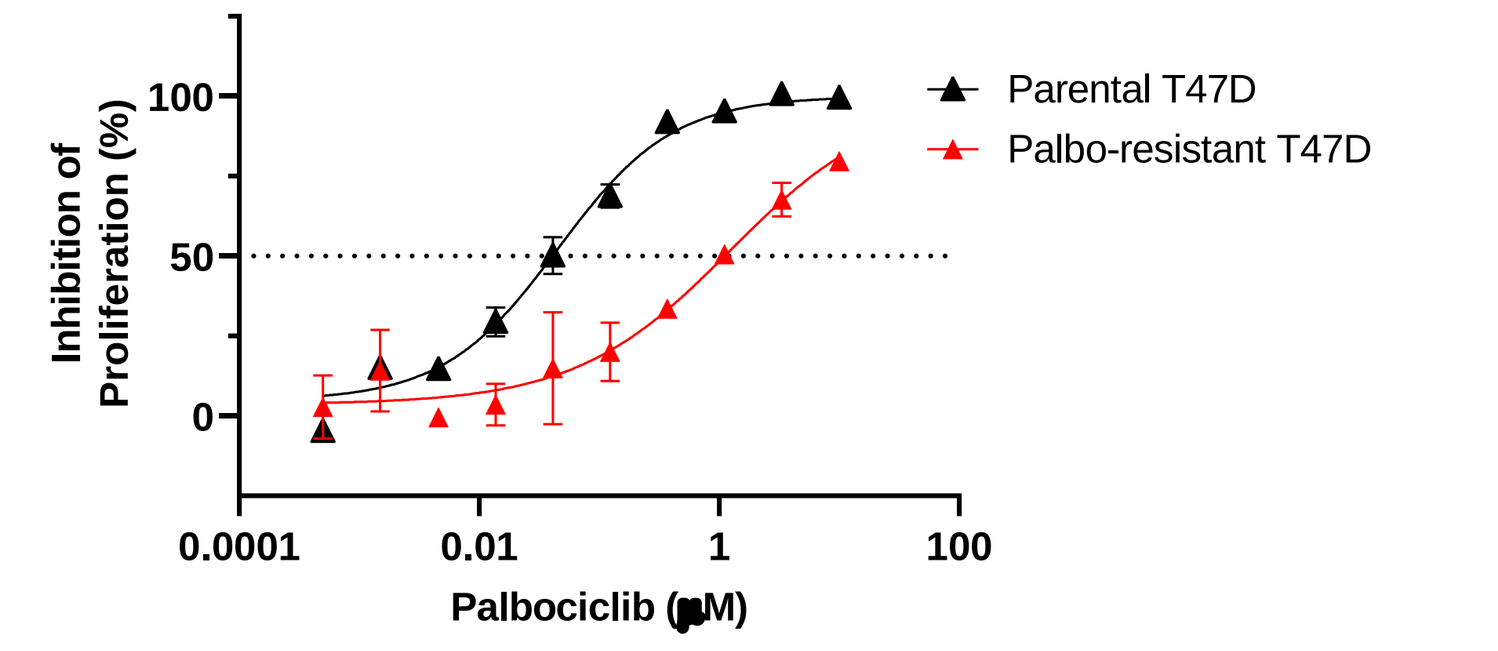
A
Inhibition of Proliferation (%)
Palbociclib (μM)
Parental T47D
Palbo-resistant T47D

	Cell	IC_{50} (μM)	Resistance fold
Palbociclib	Parental T47D	0.044	34
	Palbo-resistant T47D	1.5	

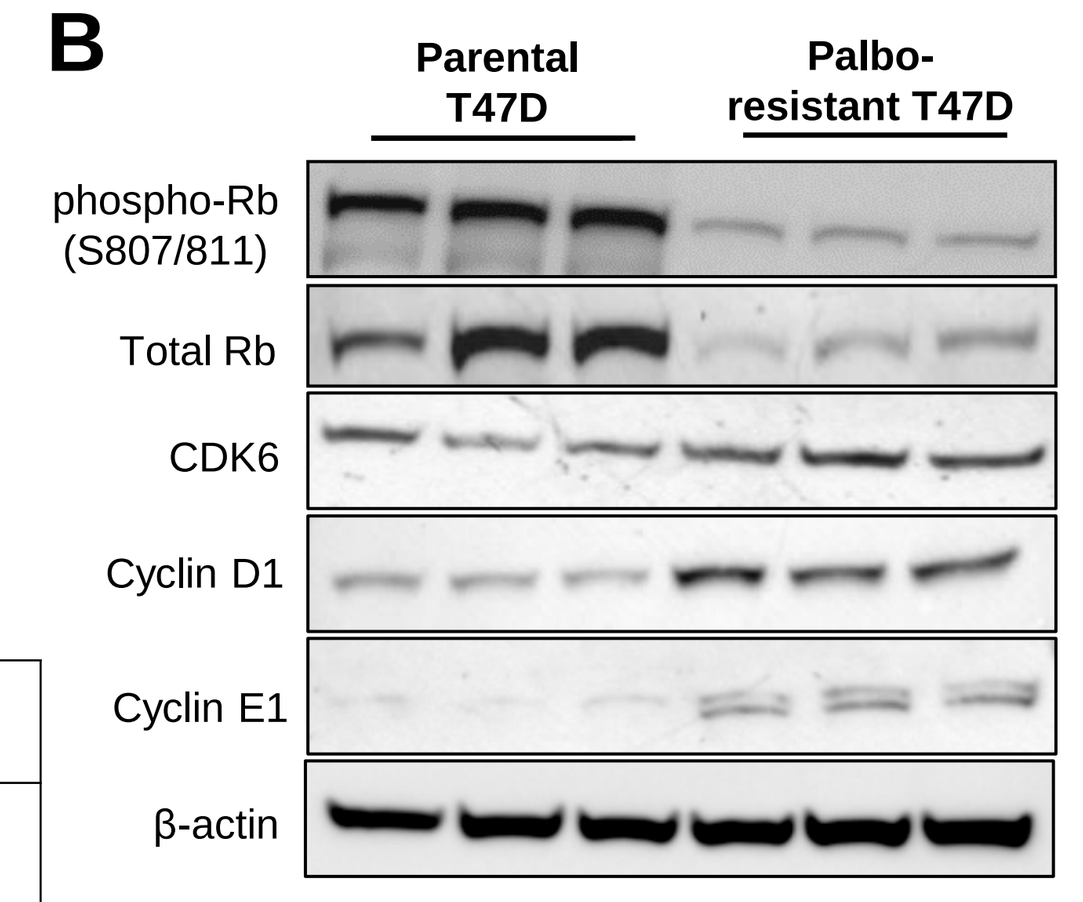
B
Parental T47D
Palbo-resistant T47D
phospho-Rb (S807/811)
Total Rb
CDK6
Cyclin D1
Cyclin E1
β-actin

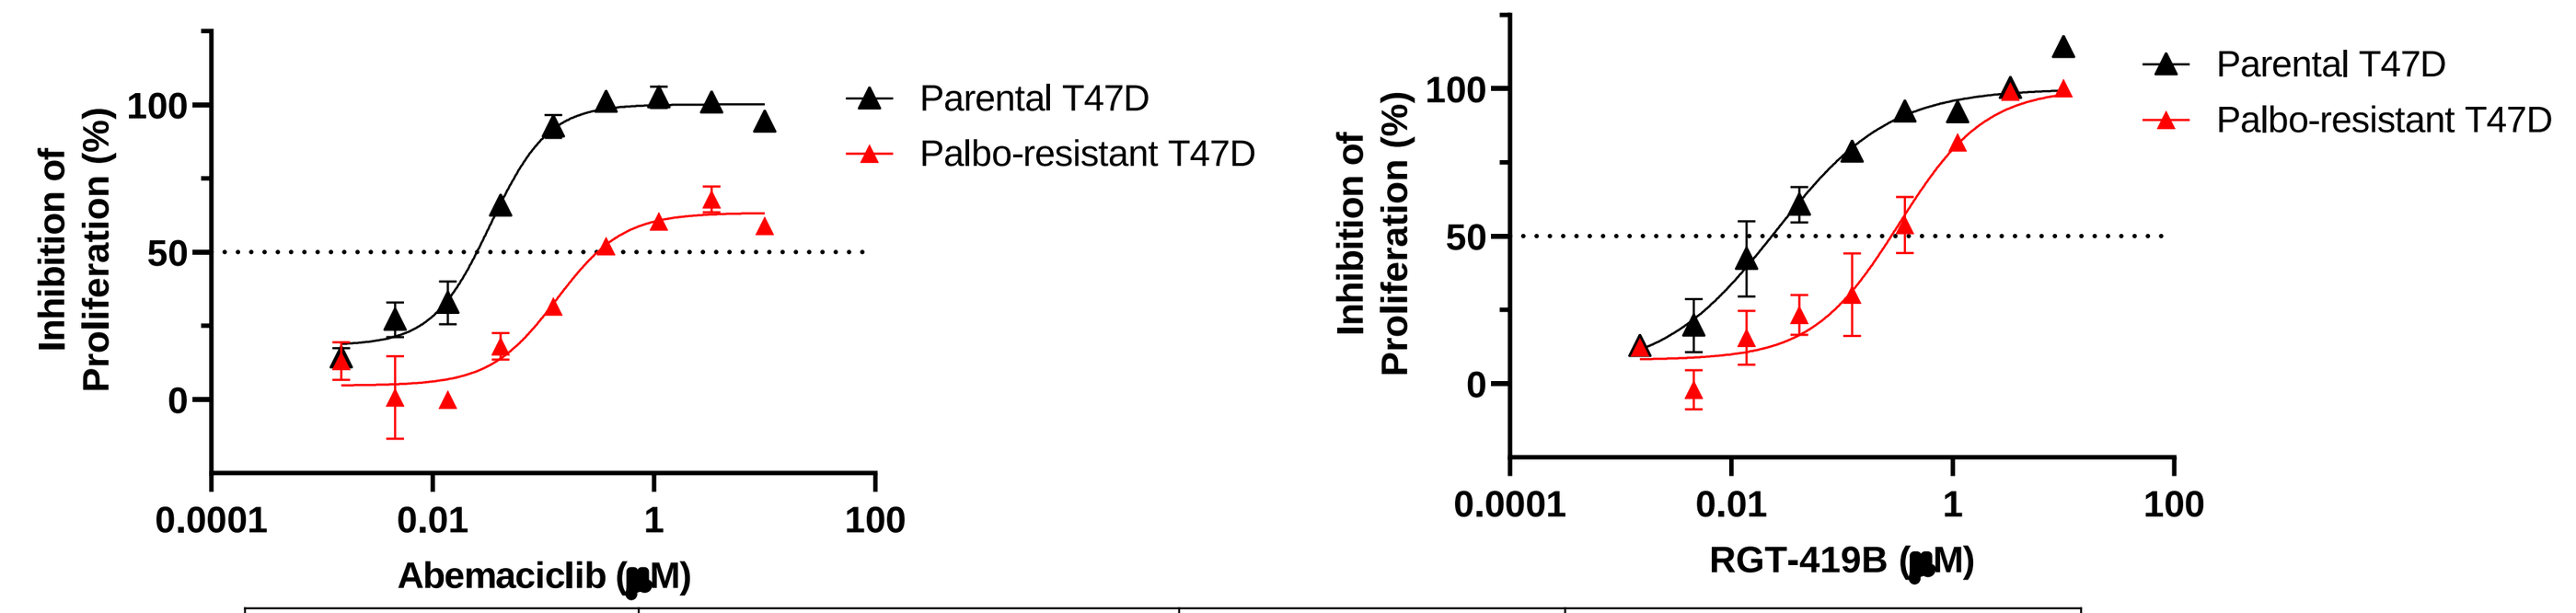
C
Inhibition of Proliferation (%)
Abemaciclib (μM)
RGT-419B (μM)
Parental T47D
Palbo-resistant T47D

	Cell	IC_{50} (μM)	Full Suppression of Palbo-resistant T47D?
Abemaciclib	Parental T47D	0.023	NO
	Palbo-resistant T47D	0.41	
RGT-419B	Parental T47D	0.032	YES
	Palbo-resistant T47D	0.31	

Figure 3. RGT-419B demonstrated full suppression of the proliferation of polyclonal Palbo-resistant T47D cells, while abemaciclib showed partial inhibition. (A) Palbo-resistant T47D cell line was established by maintaining parental T47D cells in culture for > 6 months in medium containing increasing concentrations of palbociclib. (B) Increased level of CDK6, Cyclin E1 and Cyclin D1 was observed in the polyclonal Palbo-resistant T47D cells compared to parental cells by western blot. (C) Anti-proliferation activity of abemaciclib and RGT-419B in parental and Palbo-resistant T47D cells.

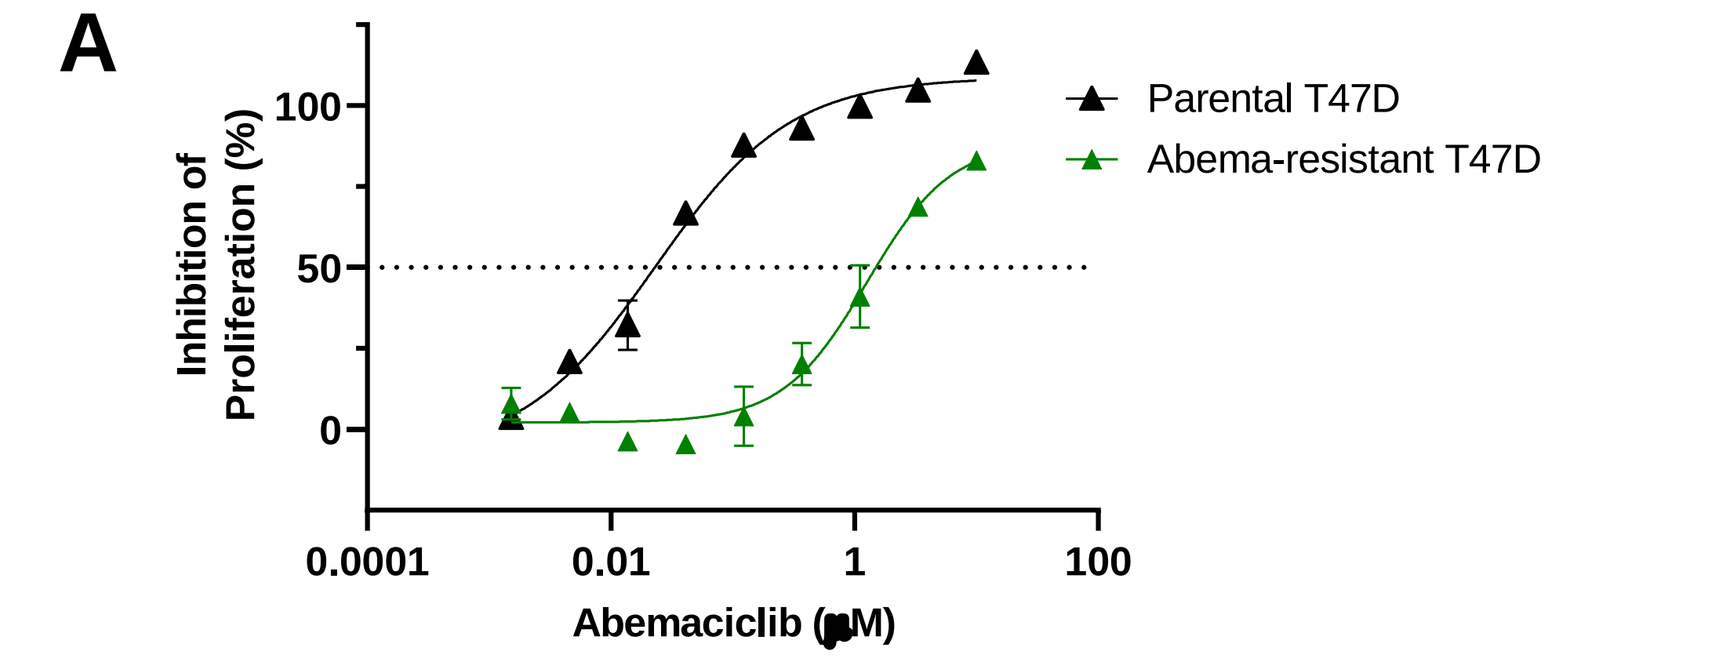
A
Inhibition of Proliferation (%)
Abemaciclib (μM)
Parental T47D
Abema-resistant T47D

	Cell	IC_{50} (μM)	Resistance fold
Abemaciclib	Parental T47D	0.019	79
	Abema-resistant T47D	1.5	

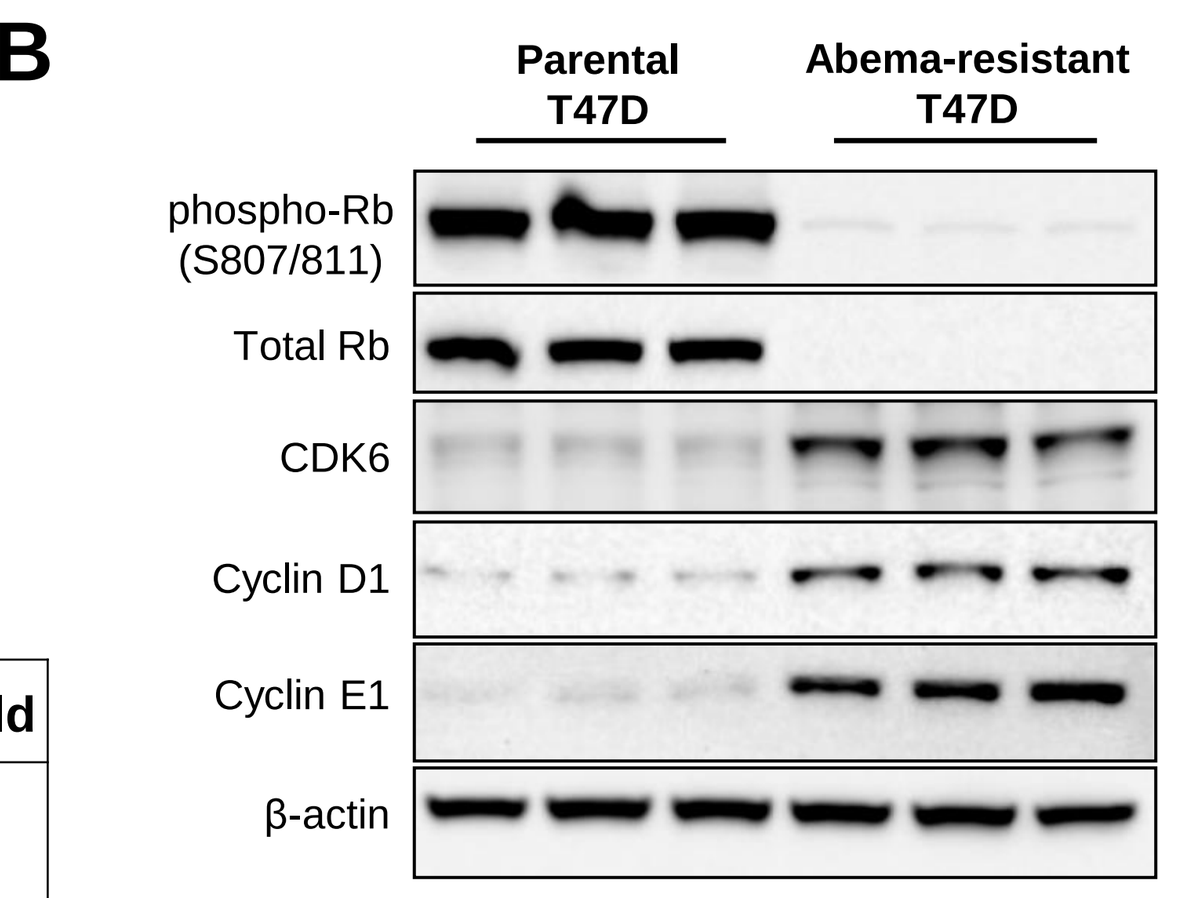
B
Parental T47D
Abema-resistant T47D
phospho-Rb (S807/811)
Total Rb
CDK6
Cyclin D1
Cyclin E1
β-actin

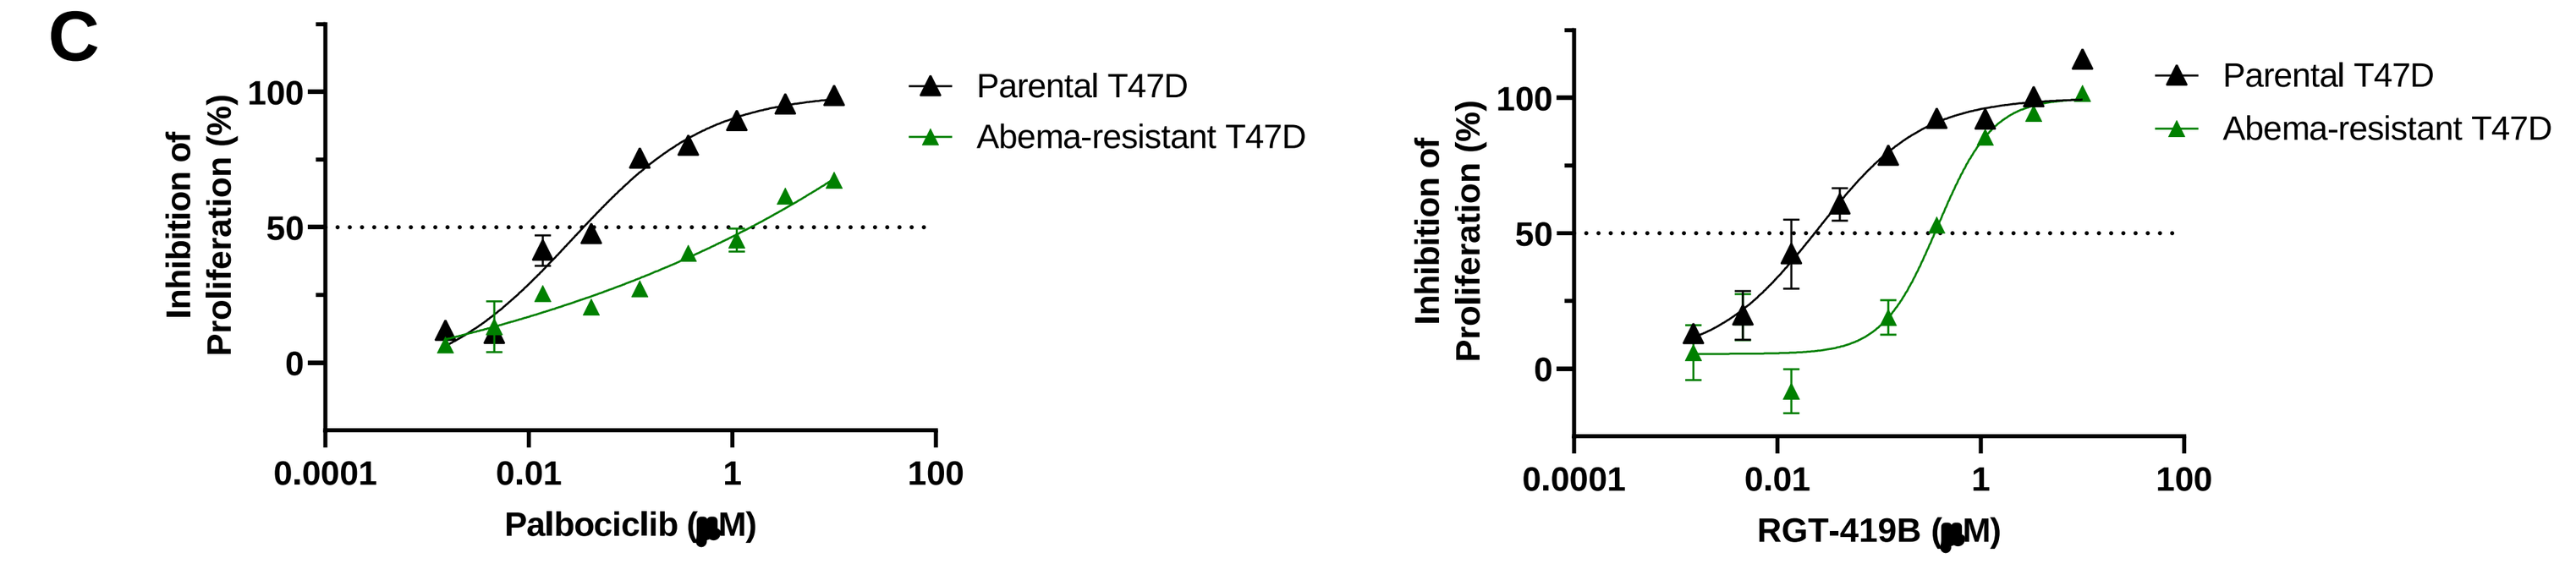
C
Inhibition of Proliferation (%)
Palbociclib (μM)
RGT-419B (μM)
Parental T47D
Abema-resistant T47D

	Cell	IC_{50} (μM)	Full Suppression of the Abema-resistant T47D cells?
Palbociclib	Parental T47D	0.043	NO
	Abema-resistant T47D	1.9	
RGT-419B	Parental T47D	0.032	YES
	Abema-resistant T47D	0.38	

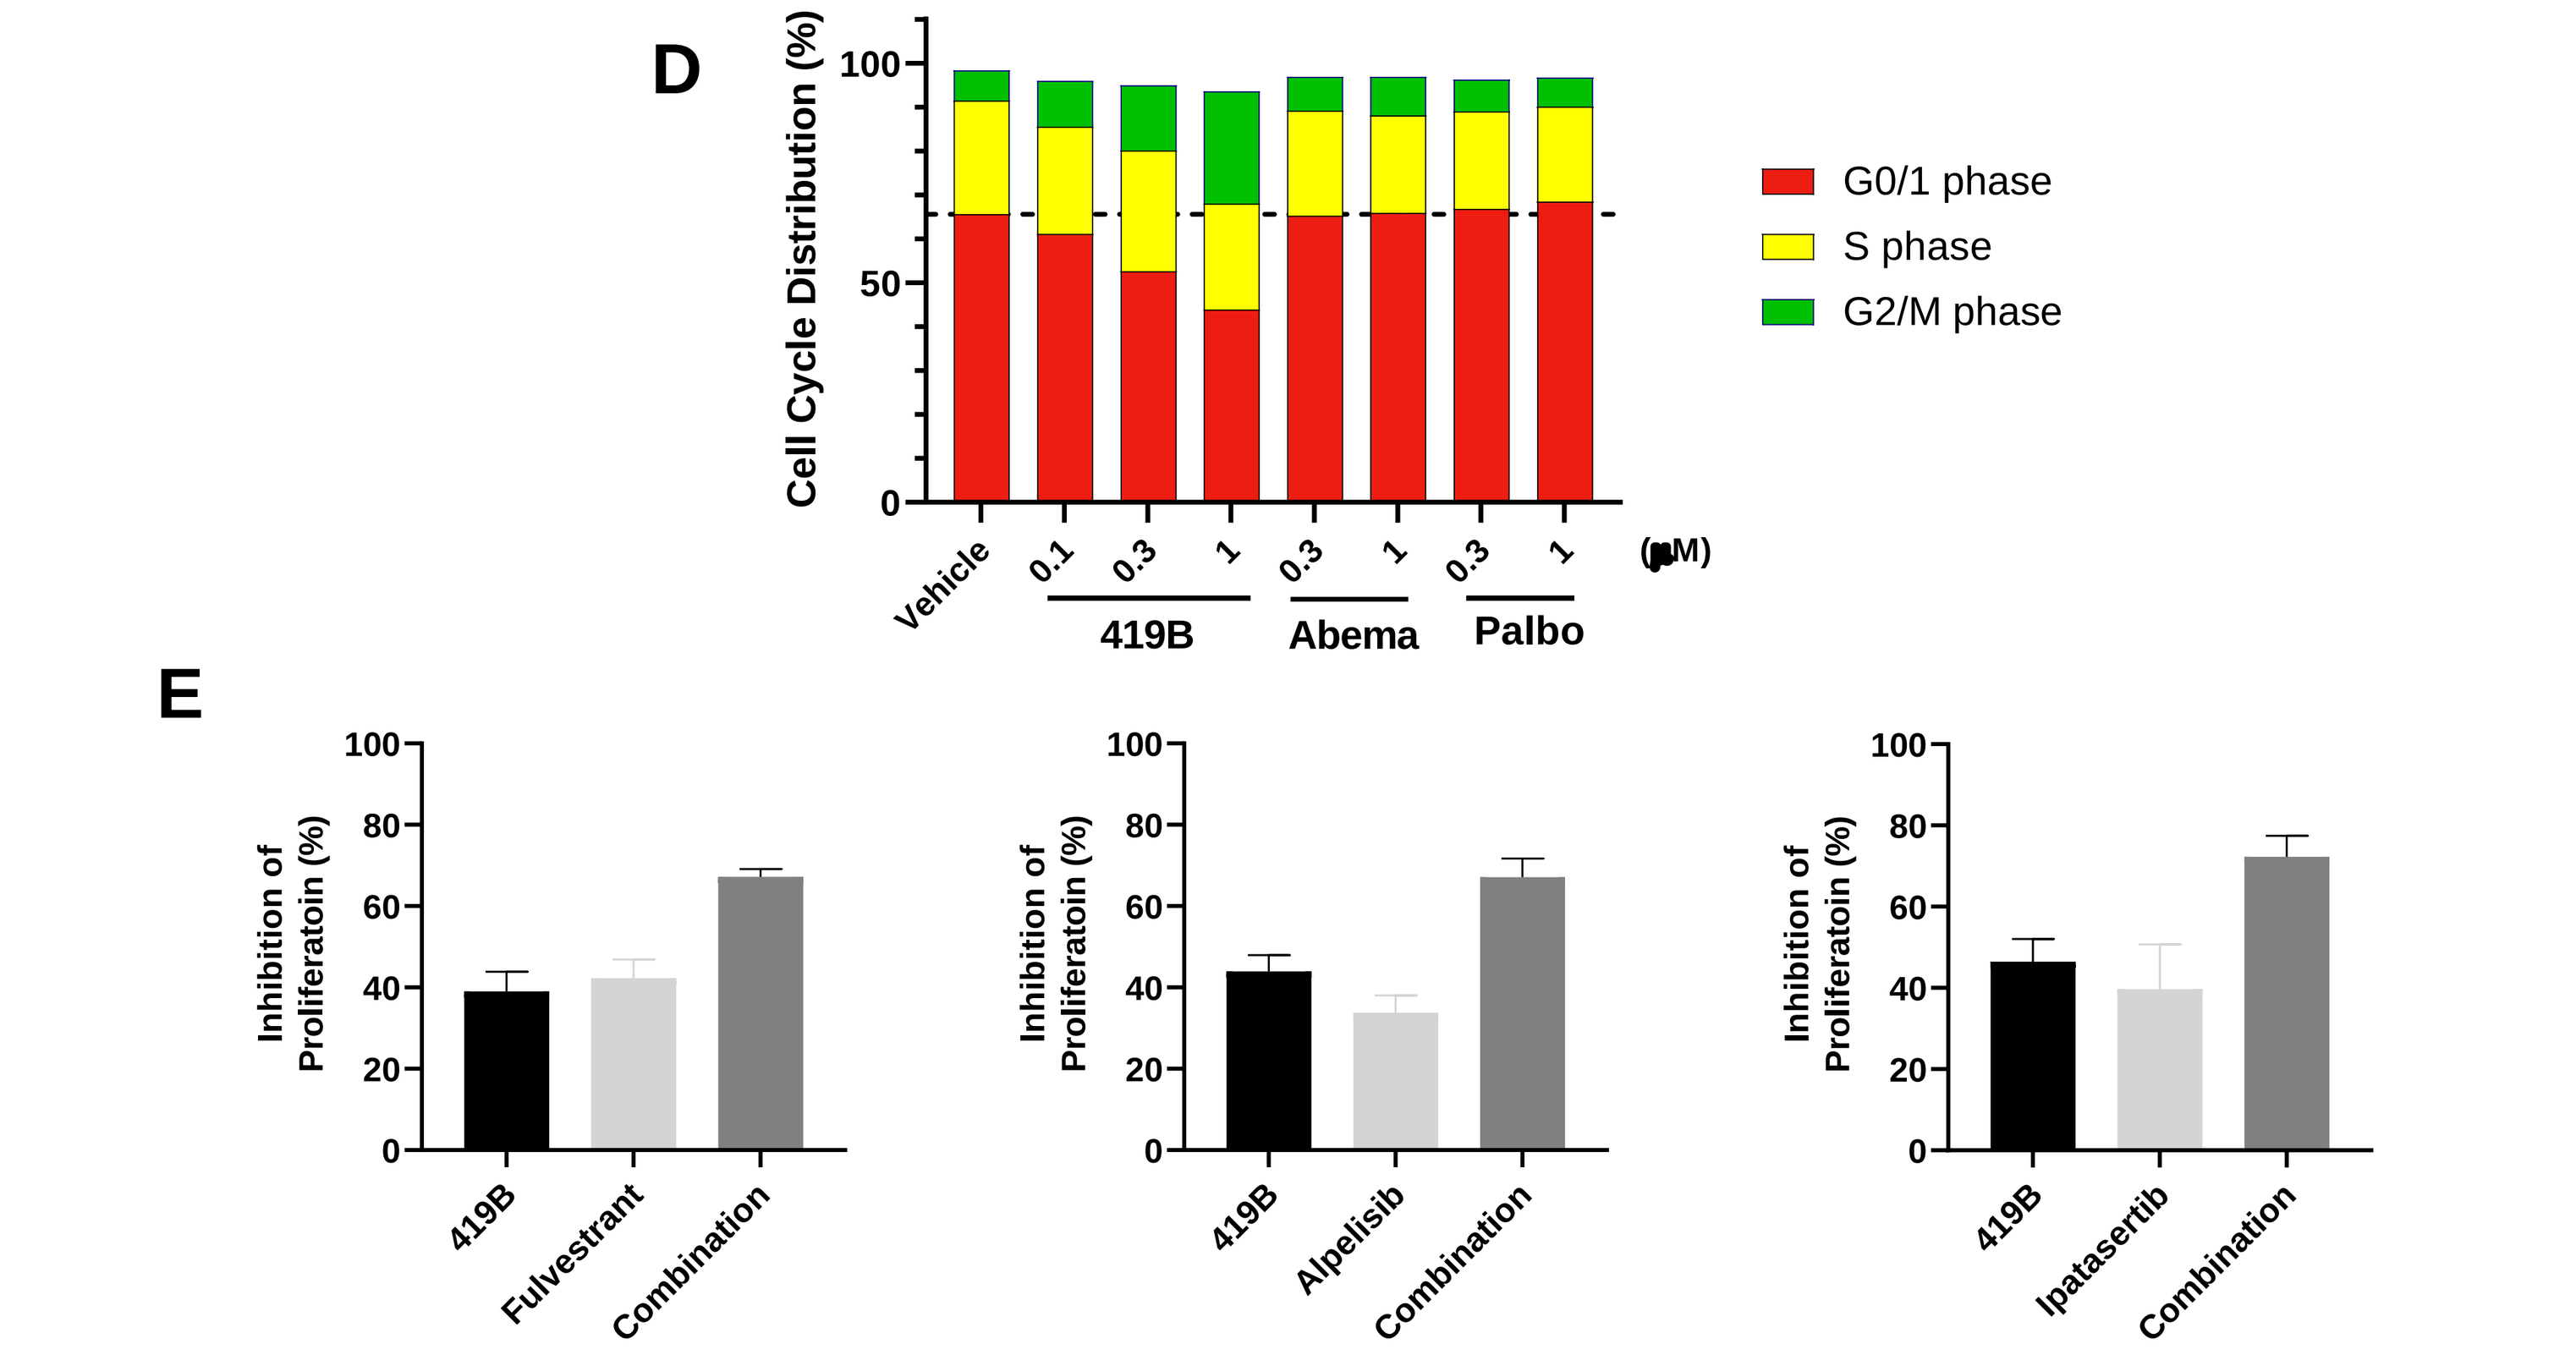
D
Cell Cycle Distribution (%)
G0/1 phase
S phase
G2/M phase
Vehicle
419B
Abema
Palbo
(μM)
E
Inhibition of Proliferatoin (%)
419B
Fulvestrant
Alpelisib
Ipatasertib
Combination

Figure 4. RGT-419B robustly inhibited the cell proliferation of the polyclonal abemaciclib-resistant T47D cells as single agent and in the combination with SERD or the inhibitors of PI3K signaling pathway. (A) Establishment of the Abema-resistant T47D cells. (B) Increased level of CDK6, Cyclin E1 and Cyclin D1 was observed in the Abema-resistant T47D cells by western blot analysis. (C) Anti-proliferation activity of palbociclib and RGT-419B in the parental and Abema-resistant T47D cells. (D) Cell cycle distribution in Abema-resistant T47D cells. (E) Combination activity of RGT-419B (0.37 μM) with fulvestrant (3 nM), alpelisib (0.3 μM, PI3Ki), or ipatasertib (0.3 μM, AKTi) in the Abema-resistant T47D cell proliferation.

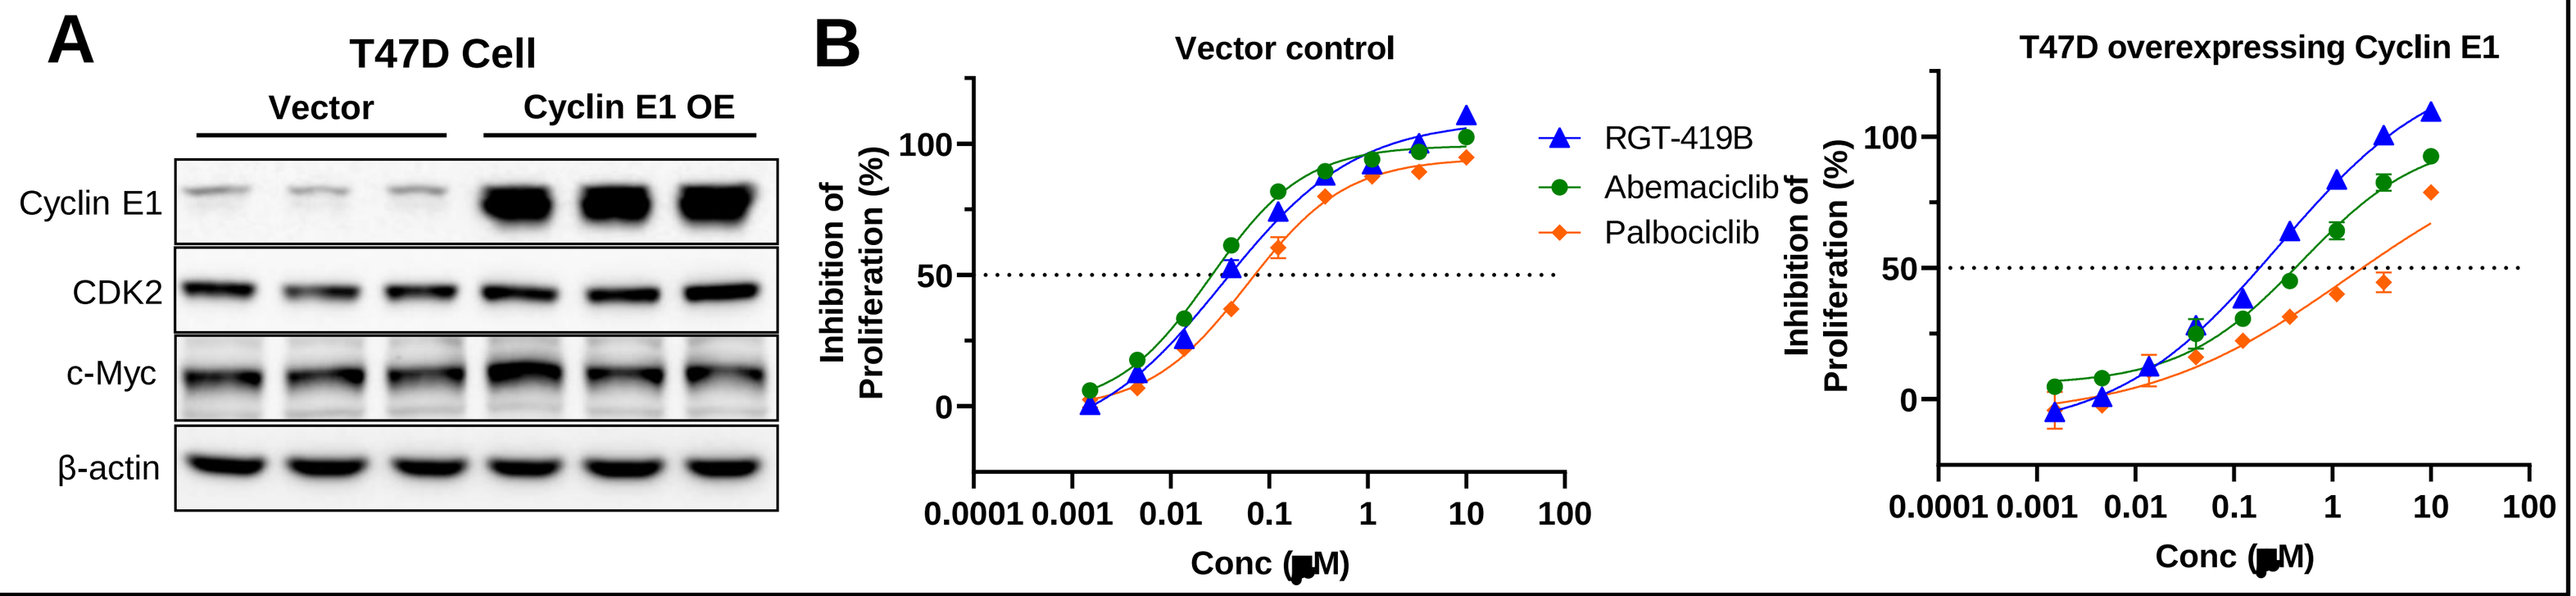
A
T47D Cell
Vector
Cyclin E1 OE
Cyclin E1
CDK2
c-Myc
β-actin
B
Vector control
T47D overexpressing Cyclin E1
Inhibition of Proliferation (%)
Conc (μM)
RGT-419B
Abemaciclib
Palbociclib

C

	Cell	IC_{50} (μM)	Folds of IC_{50} shift
RGT-419B	Vector	0.039	5
	Cyclin E1 OE	0.185	
Abemaciclib	Vector	0.025	16
	Cyclin E1 OE	0.396	
Palbociclib	Vector	0.072	27
	Cyclin E1 OE	1.951	

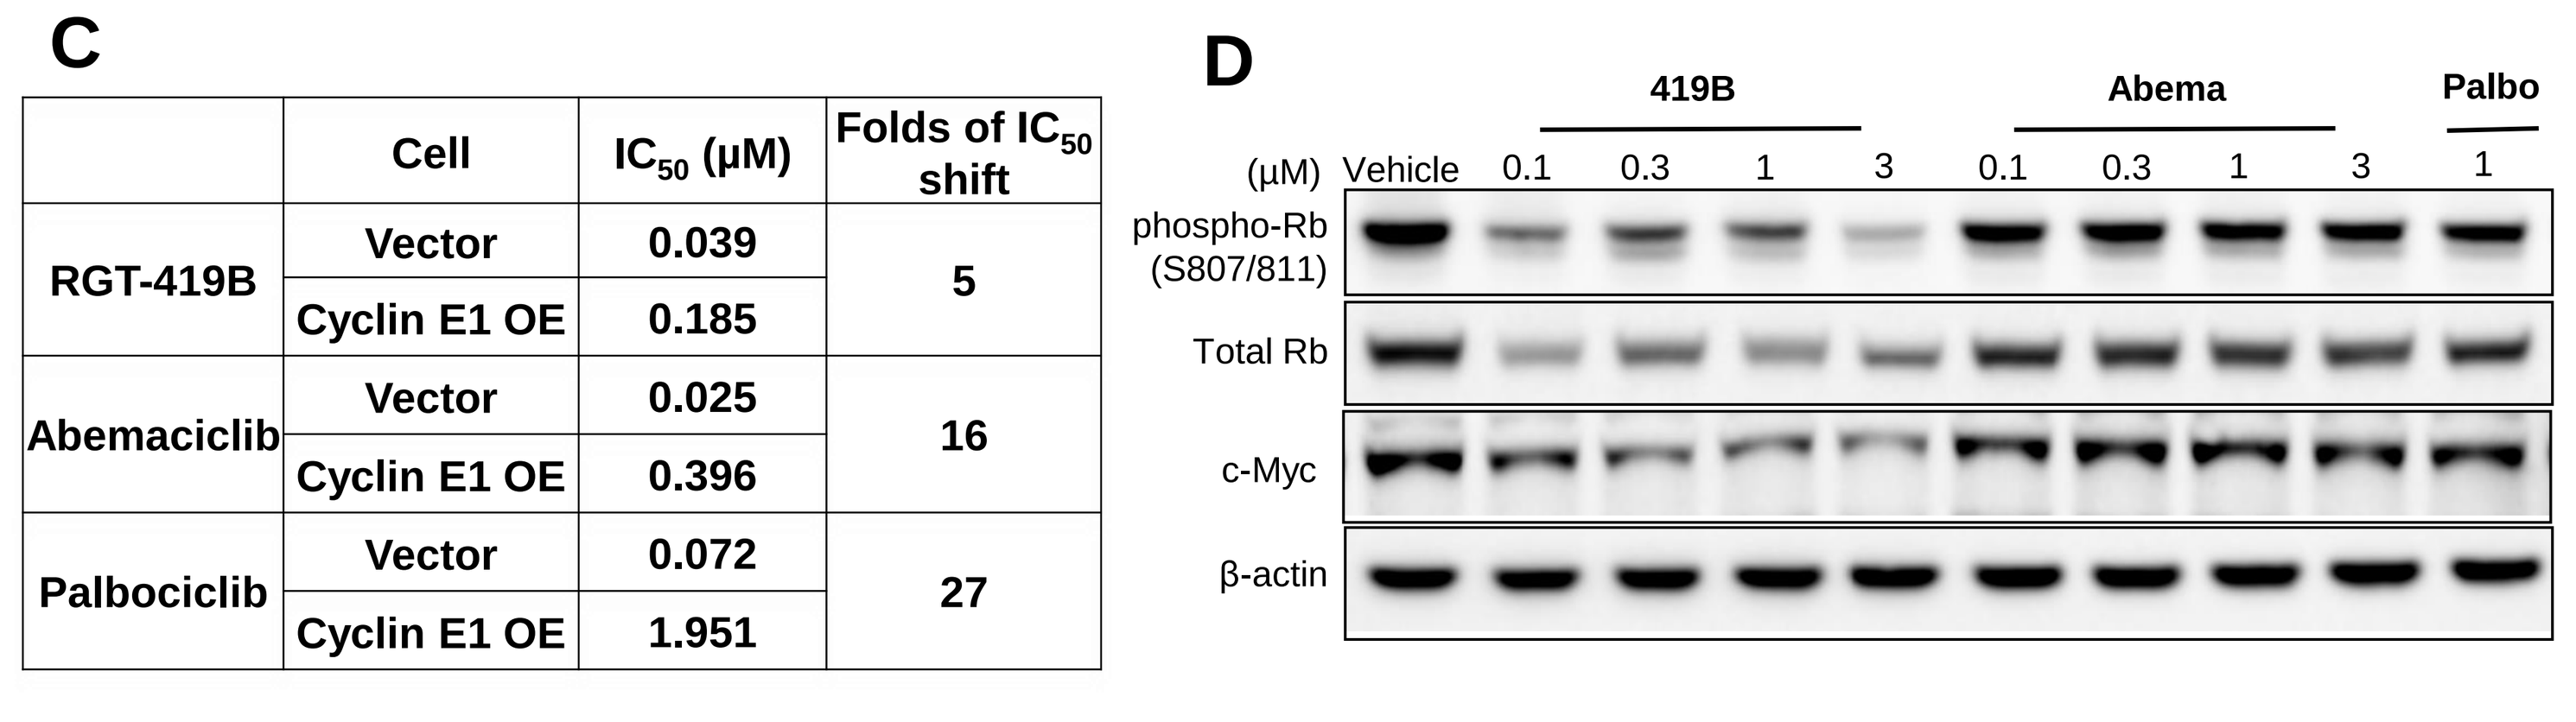
D
419B
Abema
Palbo
(μM)
Vehicle
0.1
0.3
1
3
phospho-Rb (S807/811)
Total Rb
c-Myc
β-actin

Figure 5. RGT-419B demonstrated more robust anti-proliferation activity than either abemaciclib or palbociclib in the T47D cells overexpressing Cyclin E1. (A) T47D cells overexpressing Cyclin E1 (Cyclin E1 OE) were generated by infection with recombinant lentiviruses expressing Cyclin E1 from the constitutive EF1A promoter. (B) and (C) RGT-419B exhibited more robust anti-proliferation activity than either abemaciclib or palbociclib in Cyclin E1 OE cells. (D) RGT-419B inhibited the Phospho-Rb (S807/811) and decreased the expression level of c-Myc in the Cyclin E1 OE cells.

CONCLUSIONS

- RGT-419B is a new generation CDK4/2/6 inhibitor with an optimized kinase selectivity spectrum, aiming to improve safety and combat resistance of the currently approved CDK4/6 inhibitors.
- RGT-419B caused G1 cell cycle arrest and suppressed the proliferation of ER+ T47D cells *in vitro*.
- It demonstrated sustained tumor growth inhibition in the ER+ breast cancer MCF7 xenograft as single agent *in vivo*.
- Furthermore, RGT-419B demonstrated full suppression of the proliferation of the ER+ breast cancer cells with acquired resistance to the currently approved CDK4/6 inhibitors as single agent, and the activity can be further augmented in combination with SERDs and inhibitors of PI3K signaling pathway *in vitro*.
- In ER+ T47D cells overexpressing Cyclin E1, RGT-419B exhibited more potent anti-proliferation activity when compared with abemaciclib and palbociclib.

In summary, RGT-419B, with an optimized kinase activity spectrum, provides an opportunity to treat ER+ breast cancer patients with primary or acquired resistance to the currently approved CDK4/6 inhibitors as either a single agent or in combination with other therapies such as next-Gen SERDs.

REFERENCES

1. *The Lancet, 2020, 395 (10226) 817-827*
2. *Clin Cancer Res. 2017, 23(15): 4055–4065*
3. *J Clin Oncol. 2019; 37(14): 1169–1178.*

This presentation is the intellectual property of the Regor companies. Contact zhi.xie@regor.com for permission to reprint and/or distribute.